#5667

Fatty Acid Synthase Inhibition for Ovarian Cancer.

Rajaâ El Meskini[1], Susan M. Medghalchi[1], Aravinda Vadlamudi[1], Wan Fang Han[2], Weibo Zhou[2], Yajun Tu[2], Karen Dwyer[1], Craig A. Townsend[6], Francis P. Kuhajda[2,4,5]

FASgen, Inc.[1], Baltimore, MD 21201

Departments of Pathology[2], Neuroscience[3], Oncology[4], and Biological Chemistry[5], The Johns Hopkins University School of Medicine, Baltimore, MD 21205

Department of Chemistry[6], The Johns Hopkins University Baltimore, MD 21218

Introduction

Fatty acid synthase (FAS) is highly expressed in many human cancers, including ovarian cancer. FAS is responsible for de novo synthesis of fatty acids in tumors and the overexpression of FAS in metastatic tumors correlates with decreased survival and is predictive of cancer recurrence in several tumor types. In agreement with FAS expression, FAS function is different between normal and tumor tissues. FAS functions in normal human liver and adipose tissue to store excess energy as fatty acids in the form of triglycerides, whereas in tumors, FAS functions to maintain cell energy balance. FAS is an advantageous biological target that drive tumor cell survival and proliferation and its inhibition induces tumor cell apoptosis.

Prior studies have shown that pharmacological FAS inhibition is cytotoxic to a variety of human cancer xenograft models. While successful xenograft studies suggest that FAS inhibition is not substantially toxic to normal tissues, formal toxicity data has not been described.

We have developed a most promising inhibitor of FAS, FAS 31, which is effective in vitro and in vivo against ovarian cancer cells and its mouse xenograft models. In preliminary toxicity studies, treatment with novel small molecule FAS inhibitor, FAS-31, results in no observable toxicity to normal tissues in the rat or mouse.

Methods

Cell Viability Assay: To measure the cytotoxicity of FAS-31 against cancer cells, SKOV-3 cells or OVCAR-3 were plated in 96-well plates. Following overnight culture, FAS-31 was added to the cells at specified concentrations. Vehicle controls were run for each experiment. Each condition was run in triplicate. After 72 h of incubation, cells were incubated for 4 h with the XTT reagent as per manufacturer's instructions (Cell Proliferation Kit II (XTT) Roche Diagnostics, New Jersey). IC_{50} was calculated by linear regression, plotting the FAS activity as percent of control versus drug concentrations.

Efficacy Studies: FAS-31 mouse efficacy studies were conducted in order to evaluate anti-neoplastic activity of FAS-31 in vivo. Athymic Nude mice were implanted with SKOV-3 or OVCAR-3 human ovarian cancer cell lines and were treated twice daily intraperitoneal (IP) or oral (PO) when the grown tumor xenografts reached at least 150 to 200 mm^3.

Maximum Tolerated Dose Study: Maximum Tolerated dose study was conducted to determine the toxic dose of FAS-31 in female and male rats. Sprague-Dawley rats were treated IP or PO. Animal activity, health status, food intake and body weight were monitored before and after single FAS-31 dosing.

5-Day Oral Toxicity Study: To determine the toxicity of FAS-31 in female and male, Sprague-Dawley rats were treated PO with increasing doses of FAS-31. FAS-31 was administered twice daily PO at 50 mg/Kg (as **1X** effective dose, based on mouse preliminary studies); 250 mg/Kg (**5X**), 500 mg/Kg (**10X**) to both **Females** and **Males**.

Bioavailability and Pharmacokinetics of FAS-31 in Rats and Beagle Dogs: To characterize the pharmacokinetics of FAS-31, FAS-31 was administered intra-venously (IV, at 10 mg/Kg) or orally (PO at 50 mg/Kg) in female and male, Sprague-Dawley rats or Beagle dogs. FAS-31 blood level was determined by LC-MS at different time after drug administration.

Results

Effect of FAS-31 Treatment on Human Ovarian cancer Cell lines

FAS-31 Inhibition of SKOV-3 Cell Proliferation

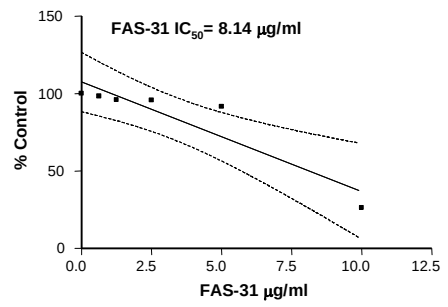
FAS-31 IC50= 8.14 µg/ml
% Control
FAS-31 µg/ml

FAS-31 Inhibition of OVCAR-3 Cell Proliferation

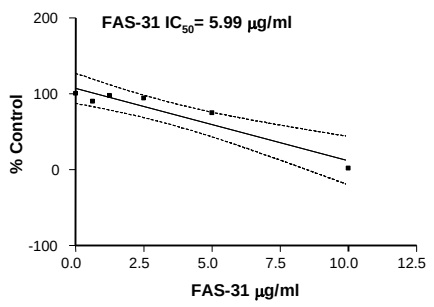
FAS-31 IC50= 5.99 µg/ml
% Control
FAS-31 µg/ml

Effect of FAS-31 Treatment on Human Ovarian cancer Xenograft model: Mouse Efficacy Studies

FAS-31 anti-neoplastic activity is specific and dose dependent. FAS-31 treatment at 50 mg/Kg twice daily intraperitoneal resulted in between 75 and 80 % reduction of the SKOV-3 tumor xenografts in 10 to 12 days period respectively. The reduction of mouse ovarian tumor volume correlates with FAS enzyme activity.

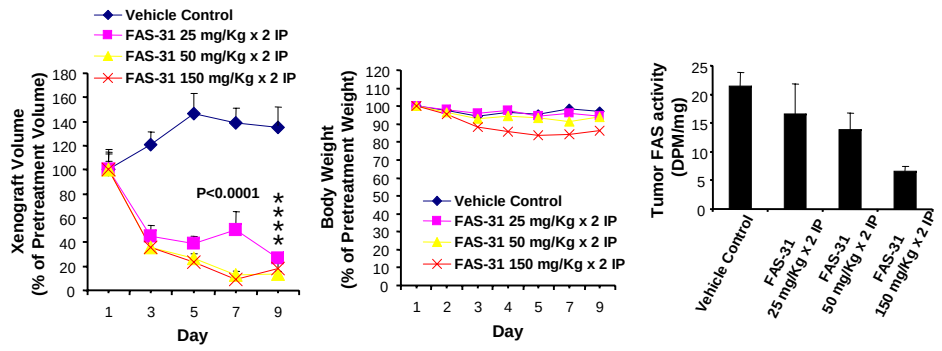
Vehicle Control
FAS-31 25 mg/Kg x 2 IP
FAS-31 50 mg/Kg x 2 IP
FAS-31 150 mg/Kg x 2 IP
Xenograft Volume (% of Pretreatment Volume)
P<0.0001
Day
Body Weight (% of Pretreatment Weight)
Tumor FAS activity (DPM/mg)
Vehicle Control
FAS-31 25 mg/Kg x 2 IP
FAS-31 50 mg/Kg x 2 IP
FAS-31 150 mg/Kg x 2 IP

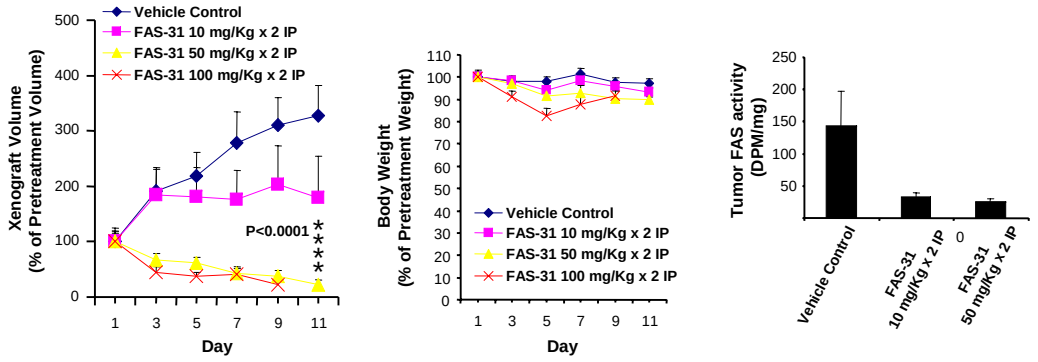
Vehicle Control
FAS-31 10 mg/Kg x 2 IP
FAS-31 50 mg/Kg x 2 IP
FAS-31 100 mg/Kg x 2 IP
Xenograft Volume (% of Pretreatment Volume)
P<0.0001
Day
Body Weight (% of Pretreatment Weight)
Tumor FAS activity (DPM/mg)
Vehicle Control
FAS-31 10 mg/Kg x 2 IP
FAS-31 50 mg/Kg x 2 IP

FAS-31 is efficacious at 50 mg/Kg via oral treatment. FAS-31 oral treatment at 50 mg/Kg twice daily resulted in almost 60% reduction of the OVCAR-3 tumor xenografts in an 11 day period. FAS-31 treatment at 50 mg/Kg twice daily resulted in almost 75% reduction of the SKOV-3 tumor xenografts in a 15 day period.

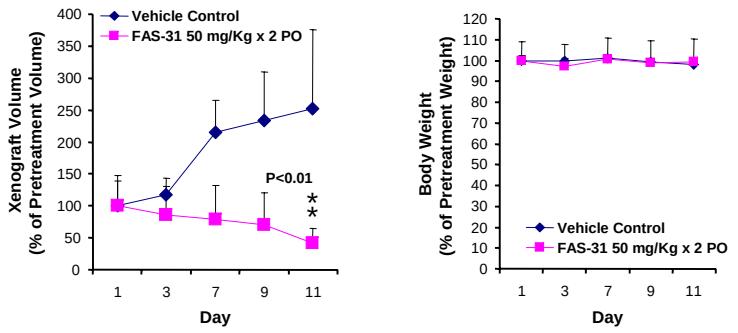
Vehicle Control
FAS-31 50 mg/Kg x 2 PO
Xenograft Volume (% of Pretreatment Volume)
P<0.01
Day
Body Weight (% of Pretreatment Weight)

OVCAR-3

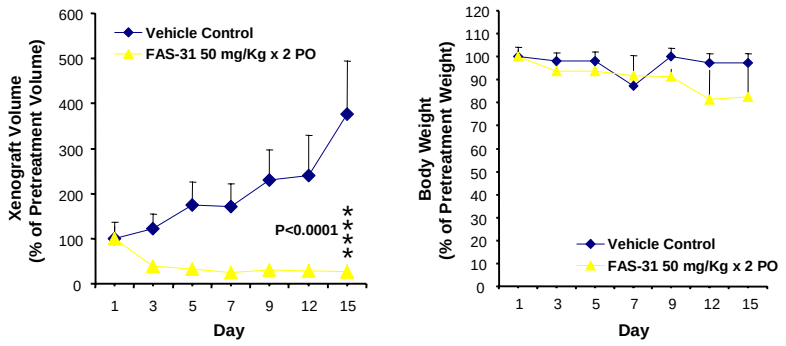
Vehicle Control
FAS-31 50 mg/Kg x 2 PO
Xenograft Volume (% of Pretreatment Volume)
P<0.0001
Day
Body Weight (% of Pretreatment Weight)

SKOV-3

FAS-31 Maximum Tolerated Dose Study in Male and Female Rats: IP and PO Route (2 representative studies)

Overall, no major toxicity was found and normal motor activity was recorded following acute FAS-31 treatment (IP) in female rats and (PO) in male rats.

Female Rat: IP Route

FAS-31 Dose (mg/KG)	Number of Animals per Group	Rat Death	Body Weight ± SEM (% of pretreatment weight)					
			Day 1	Day 2	Day 3	Day 4	Day 5	Day 8
0	3	0	100 ± 0.66	96.52 ± 1.63	94.17 ± 1.14	98.22 ± 1.48	103.10 ± 1.60	114.57 ± 2.22
50	3	0	100 ± 2.22	102.06 ± 1.81	105.89 ± 1.97	108.35 ± 2.14	110.90 ± 2.30	116.11 ± 3.59
150	3	0	100 ± 4.57	92.94 ± 3.86	95.84 ± 4.22	100.29 ± 4.48	105.80 ± 5.39	112.09 ± 7.10
300	3	0	100 ± 0.36	92.71 ± 0.67	96.23 ± 2.10	100.10 ± 1.34	106.06 ± 1.03	113.51 ± 1.47
500	3	0	100 ± 2.80	95.84 ± 4.72	96.02 ± 7.21	98.96 ± 6.60	104.07 ± 5.09	112.58 ± 4.99
1000	3	0	100 ± 3.46	96.89 ± 4.82	93.60 ± 6.22	95.95 ± 7.93	98.96 ± 7.68	110.64 ± 5.15
1500	3	1	100 ± 3.19	99.60 ± 2.10	94.38 ± 1.12	94.38 ± 3.59	94.43 ± 6.28	106.17 ± 0.62

Male Rat: PO Route

FAS-31 Dose (mg/KG)	Number of Animals per Group	Rat Death	Body Weight ± SEM (% of pretreatment weight)					
			Day 1	Day 2	Day 3	Day 6	Day 8	Day 9
0	3	0	100 ± 6.10	96.25 ± 6.35	99.18 ± 7.40	106.37 ± 7.60	110.12 ± 7.64	110.66 ± 7.49
50	3	0	100 ± 4.72	105.57 ± 1.84	102.21 ± 5.48	107.94 ± 6.13	112.30 ± 7.02	104.26 ± 4.50
150	3	0	100 ± 1.10	98.39 ± 0.64	99.54 ± 0.69	106.49 ± 0.21	109.53 ± 0.69	107.92 ± 0.11
300	3	0	100 ± 1.88	99.46 ± 2.88	101.55 ± 2.88	105.94 ± 2.70	108.73 ± 2.79	107.34 ± 2.74
500	3	0	100 ± 3.12	96.08 ± 3.86	100.60 ± 4.53	108.22 ± 4.28	110.89 ± 4.51	109.69 ± 4.37
1000	3	0	100 ± 3.64	99.22 ± 4.37	99.16 ± 4.25	105.70 ± 5.28	108.78 ± 5.18	107.48 ± 6.08
1500	3	0	100 ± 6.37	98.63 ± 5.95	97.53 ± 1.12	105.05 ± 7.02	107.57 ± 7.19	107.41 ± 7.38

FAS-31 5 day Toxicity Study in Male and Female Rat: PO Route

No abnormal behavior or distress was associated with FAS-31 treatment twice daily in female and male rats.

Male Rat: PO Route

FAS-31 Dose (mg/KG)	Number of Animals per Group	Body Weight ± SEM (% of pretreatment weight)					Average Food Intake (g/g of Body Weight X 10^{-2})			
		Day 1	Day 2	Day 3	Day 4	Day 5	Day 2	Day 3	Day 4	Day 5
0 (0.25 ml/Kg)	5	100 ± 2.38	98.88 ± 3.91	102.24 ± 3.78	106.61 ± 4.04	110.64 ± 3.89	28.04	30.39	31.72	26.85
0 (2.5 ml/Kg)	5	100 ± 1.64	91.87 ± 2.22	93.04 ± 4.78	102.20 ± 1.10	104.40 ± 1.10	11.74	9.64	13.20	18.01
50	5	100 ± 2.07	98.83 ± 2.59	102.04 ± 2.59	107.30 ± 1.78	113.43 ± 2.84	23.25	30.11	27.12	31.82
250	5	100 ± 2.56	96.29 ± 3.91	96.63 ± 3.39	101.69 ± 3.93	106.41 ± 4.15	19.20	21.94	25.39	27.78
500	5	100 ± 2.23	97.91 ± 3.11	99.81 ± 2.69	100.00± 4.44	106.33 ± 3.85	15.18	17.95	16.40	25.25

Female Rat: PO Route

FAS-31 Dose (mg/KG)	Number of Animals per Group	Body Weight ± SEM (% of pretreatment weight)					Average Food Intake (g/g of Body Weight X 10^{-2})			
		Day 1	Day 2	Day 3	Day 4	Day 5	Day 2	Day 3	Day 4	Day 5
0 (0.25 ml/Kg)	5	100 ± 3.19	103.41 ± 3.12	104.80 ± 2.74	105.44 ± 3.21	105.31 ± 3.78	37.35	28.20	22.42	22.42
0 (2.5 ml/Kg)	5	100 ± 3.88	96.57 ± 4.19	98.16 ± 4.04	100.00 ± 4.41	102.50 ± 4.79	12.60	19.74	24.35	23.74
50	5	100 ± 2.98	100.13 ± 3.66	101.16 ± 5.16	106.64 ± 3.67	108.41 ± 4.17	19.37	27.41	23.80	21.93
250	5	100 ± 3.61	103.42 ± 3.61	106.84 ± 2.62	110.31 ± 3.00	113.57 ± 3.05	13.91	14.36	18.96	16.40
500	5	100 ± 1.69	102.42 ± 2.23	99.19 ± 1.86	101.48 ± 3.27	104.44 ± 4.14	13.66	11.47	19.15	18.24

Pharmacokinetics of Geometric Mean Plasma Concentration in Rat Following FAS-31 Oral Administration of 50 mg/Kg

Rat/Plasma	Male	Female
FAS-31/F1	PO (50 mg/Kg)	PO (50 mg/Kg)
Half Life (h) in Plasma	5.07	3.8
Half Life (h) in Lung	3.55	4.38
Volume of Distribution ± SD (Vd, L/Kg)	62.38 ± 9.08	66.53 ± 6.12
AUC ± SD (ng/ml x h) in Plasma	5982.7 ± 1160.61	4133.7 ± 458
Cmax (ng/ml)	750.2 ± 102.25	687.15 ± 57.75
AUC ± SD (ng/ml x h) in Lung	14102.98 ± 2906.57	11287.8 ± 2323.35

Pharmacokinetics of Geometric Mean Plasma Concentration in Beagle Dog Following FAS-31 Intravenous Infusion of 10 mg/Kg or Oral Administration of 50 mg/Kg

Formulation	FAS31/F1/F2	FAS-31/F1		FAS-31/F2	
Dog/Plasma	Male/Female	3 Male/2 Female	3 Male/2 Female	3 Male/3 Female	3 Male/3 Female
State	IV (10 mg/Kg)	Fasted	Fed	Fasted	Fed
		PO (50 mg/Kg)	PO (50 mg/Kg)	PO (50 mg/Kg)	PO (50 mg/Kg)
Half Life (h) in Plasma	1.75	2.42	3.6	2.58	2.9
Volume of Distribution ± SD (Vd, L/Kg)	V1= 1.63 ± 0.11 V2= 4.26 ± 0.91	V1= 73.21 ± 10.42 V2= 15.76 ± 7.22	V1= 21.98 ± 2.17 V2= 8.50 ± 5.10	V1= 98.91 ± 7.88 V2= 20.26 ± 12.92	V1= 31.14 ± 4.36 V2= 8.25 ± 4.58
AUC ± SD (ng/ml x h) in Plasma	1695.5 ± 78.43	1431.39 ± 30.96	4352.14 ± 296.29	917.60 ± 47.80	4825.48 ± 272.03
Cmax (ng/ml)	3485.13 ± 153.47	435.58 ± 10.99	1702.66 ± 119.29	435.14 ± 27.65	1339.69 ± 95.27
Bioavailability (%) in Plasma		16.88	51.33	10.82	56.92

CONCLUSIONS

- **FAS-31 is efficacious against ovarian cancer tumors.**
- **FAS-31 dosing achieved in the toxicological studies is sufficient to induce apoptosis in human cancer cells and is non toxic.**
- **FAS-31 is a viable pharmacological target for anti-cancer therapy.**